Remibrutinib versus placebo in CSU patients: REMIX-1 and REMIX-2 trials

Michihiro Hide,[1] Marcus Maurer,[2] Ana Giménez-Arnau,[3] Sarbjit Saini,[4] Mark Lebwohl,[5] Gordon Sussman,[6] El-Djouher Martzloff,[7] Karine Lheritier,[7] Artem Zharkov,[7] Sibylle Haemmerle[7]

[1]Department of Dermatology, Hiroshima Citizens Hospital, Hiroshima, Japan; [2]UCARE, Institute of Allergology, Charité – Universitätsmedizin Berlin, Germany; and Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Allergology and Immunology, Berlin, Germany; [3]Department of Dermatology, Hospital del Mar -IMIM, Universitat Pompeu Fabra, Barcelona, Spain; [4]Clinical Research Center, Little Rock Allergy and Asthma Clinic, Little Rock Arkansas, USA; [5]Department of Dermatology, Icahn School of Medicine at Mt. Sinai, New York, USA; [6]Division of Allergy and Clinical Immunology, University of Toronto, Toronto, Canada; [7]Novartis Pharma AG, Basel, Switzerland

INTRODUCTION

- Chronic spontaneous urticaria (CSU) is characterised by occurrence of itchy hives, angioedema, or both for ≥6 weeks, in the absence of specific external stimuli[1]
- More than 50% of patients respond inadequately to second-generation H_1-antihistamines treatment; thus, there is an unmet medical need for effective and safe treatment options for patients with CSU[2]
- Bruton's Tyrosine Kinase (BTK) inhibition, a promising therapeutic approach, impairs signalling downstream of FcεRI, which induces mast cell activation[3] and is being explored for the treatment of CSU[4]
- Remibrutinib (LOU064) is a novel, highly selective and potent, oral covalent BTK inhibitor (BTKi).[3] It has demonstrated clinical efficacy, fast onset of action and a favourable safety profile in CSU patients, inadequately controlled by second-generation H_1-antihistamines, in the Phase 2b dose-finding clinical study[5]

OBJECTIVE

- To present the study design of REMIX-1 (NCT05030311) and REMIX-2 (NCT05032157) – two ongoing Phase 3 clinical trials of remibrutinib in CSU patients inadequately controlled by second-generation H_1-antihistamines

METHODS

Study design

- REMIX-1 and REMIX-2 are global, multicentre, randomised, double-blind, parallel-group, placebo-controlled, Phase 3 clinical trials of near identical design. The trials are being conducted in parallel to evaluate the efficacy, safety, and tolerability of remibrutinib versus placebo in adult CSU patients inadequately controlled by second-generation H_1-antihistamines
- Patients are being randomised in the ratio of 2:1 to receive remibrutinib (n=300) or placebo (n=150). The studies consist of the following four periods with a total duration of up to 60 weeks (**Figure 1**):
 - Screening period: up to 4 weeks
 - Double-blind treatment period: 24 weeks of remibrutinib or placebo
 - Open-label treatment period: 28 weeks with remibrutinib
 - Follow-up period or extension study: 4 weeks of treatment-free follow-up or participation in an extension study

Figure 1. Study design of REMIX trials

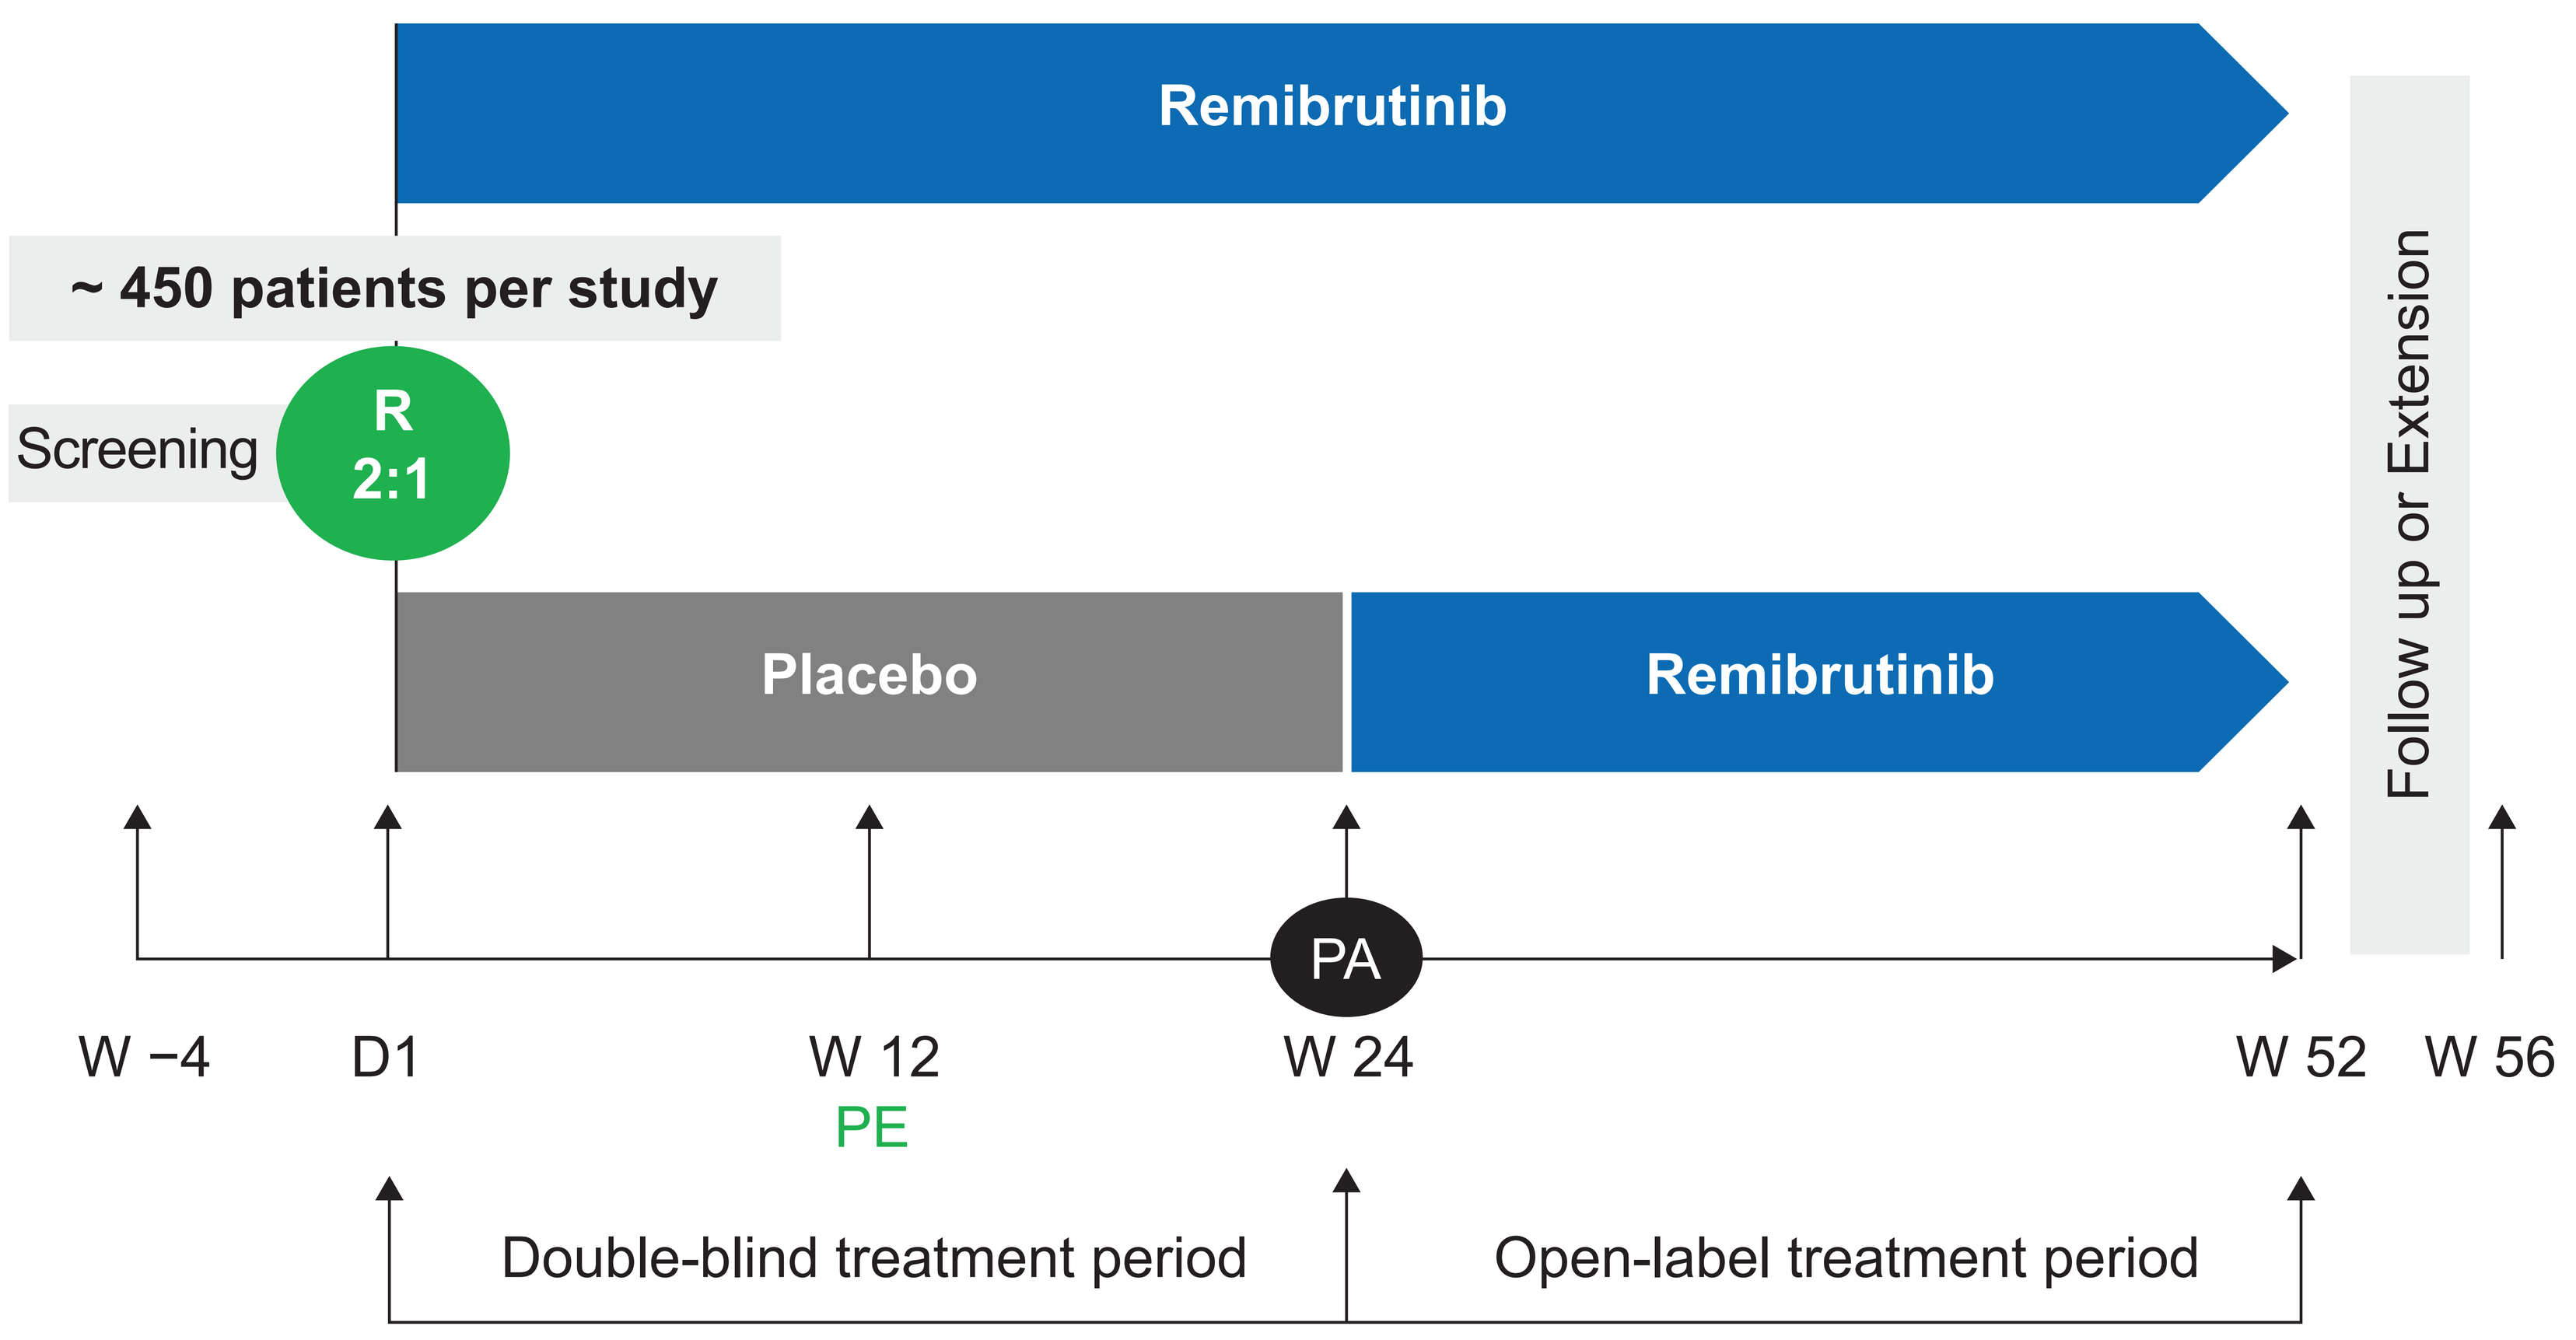
Remibrutinib
~ 450 patients per study
Screening
R
2:1
Placebo
Remibrutinib
Follow up or Extension
PA
W −4
D1
W 12
PE
W 24
W 52
W 56
Double-blind treatment period
Open-label treatment period

Participants will take background medication (second-generation H_1-antihistamines at locally label approved doses) with a stable regimen during the studies

D1, Day 1; PA, primary analysis; PE, primary endpoint; R, randomisation; W, week

- Participants will be stratified based on prior exposure to anti-IgE biologics for CSU and geographic region
- REMIX-1 trial is being conducted in 144 sites (18 countries) and REMIX-2 trial in 147 sites (18 countries) (**Figure 2**)

Figure 2. Participating countries (REMIX-1 and REMIX-2)

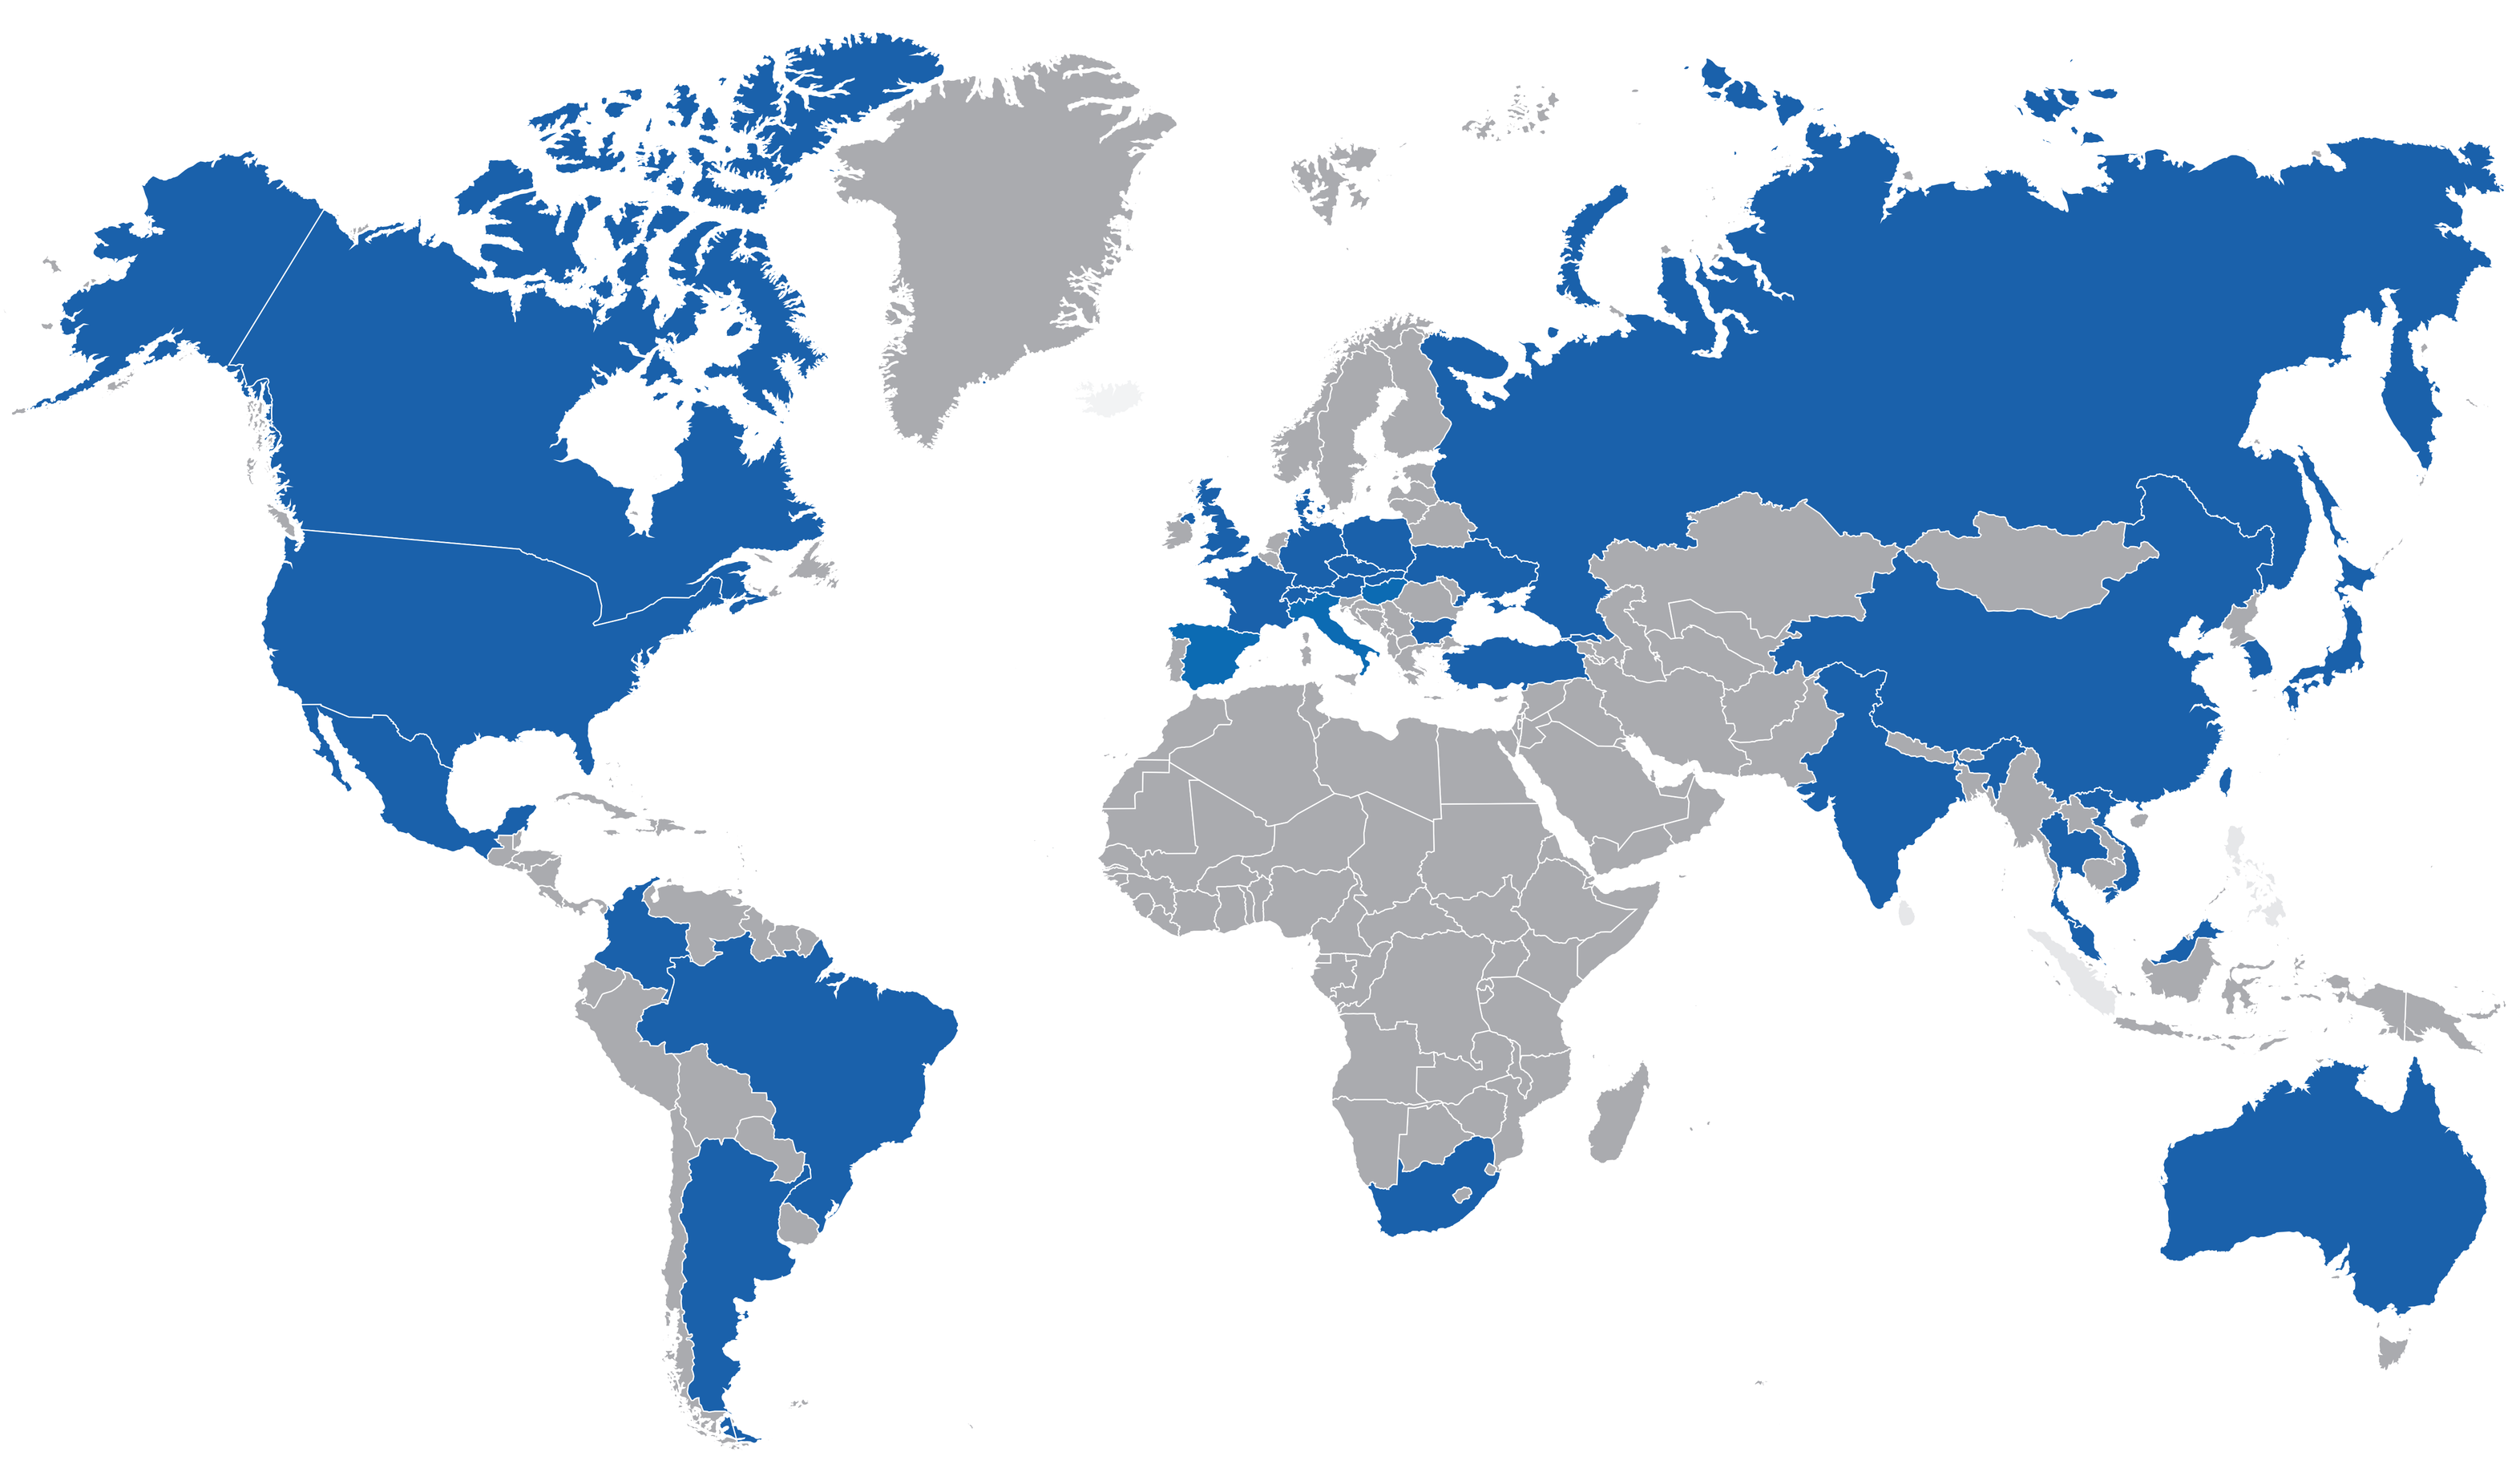

Patient population

- The study population consists of approximately 450 adult patients (in each REMIX trial) with CSU inadequately controlled by a second-generation H_1-antihistamines
- Maximum number of participants with prior exposure to anti-IgE biologics for CSU will be limited to approximately 30% of the total study population (in each REMIX trial)
- The key inclusion and exclusion criteria of the participants are listed in **Table 1**

Study objectives and endpoints

- The studies have two primary objective scenarios (depending on regional precedent and Health Authority feedback), which will be analysed independently:
 - Scenario with weekly Urticaria Activity Score (UAS7) as the primary efficacy endpoint
 - Scenario with weekly Itch Severity Score (ISS7) and weekly Hives Severity Score (HSS7) as the co-primary efficacy endpoints
- The primary and main secondary endpoints of the trials are listed in **Table 2**

Sample size determination and statistical analysis

- In order to fulfil registration and International Council for Harmonisation E1 requirements on the number of participants treated for 6 and 12 months in the development program, a sample size of 300 participants in the active arm and 150 in the placebo arm is targeted. Hence, the total sample size is 450 randomised participants
- Assuming a dropout rate of ~10% by Week 12, 270 participants in the remibrutinib arm and 135 participants in the placebo arm will provide a nominal power of >99% to detect a difference between the treatment arms
- The absolute change from baseline in UAS7 at Week 12 will be analysed using Mixed Model Repeated Measure (MMRM) modelling including treatment group, baseline UAS7 score, randomisation strata variables, week and both interaction of treatment by week and interaction of baseline UAS7 score by week as fixed effects
- Similarly, the absolute change from baseline in ISS7 and HSS7 at Week 12 will be analysed using MMRM modelling including treatment group, baseline ISS7 score (respectively HSS7), randomisation strata variables, week and both interaction of treatment by week and interaction of baseline ISS7 score (respectively HSS7) by week as fixed effects
- Disease activity control (% of participants achieving UAS7≤6) at Week 12, and complete absence of hives and itch at Week 12 (% of participants achieving UAS7=0) will be analysed using a logistic regression model, including treatment group, randomisation strata variables, and baseline UAS7 score as covariates
- All the efficacy analyses will be conducted on the full analysis set who received study treatment by randomisation. All safety variables will be conducted in the safety set, defined as participants who received at least one dose of study treatment
- The analyses of primary and secondary endpoints are planned to use the multiple testing strategy to control the family-wise error at α=0.025 (one-sided)

Table 1. Key inclusion and exclusion criteria

Key inclusion criteria
• Male and female adult participants (≥18 years) • CSU duration for ≥6 months prior to screening • Diagnosis of CSU inadequately controlled by second-generation H_1-antihistamines at the time of randomisation defined as: – Presence of itch and hives for ≥6 consecutive weeks prior to screening despite the use of a second-generation H_1-antihistamines during this time period – UAS7 score (range 0–42) ≥16, ISS7 score (range 0–21) ≥6 and HSS7 score (range 0–21) ≥6 during the 7 days prior to randomisation (Day 1) • Documentation of hives within three months before randomisation
Key exclusion criteria
• A clearly defined predominant or sole trigger of their chronic urticaria (CIndU) • Other diseases that present with wheals or angioedema • Any other skin disease associated with chronic itching • Evidence of clinically significant cardiovascular, neurological, psychiatric, pulmonary, renal, hepatic, endocrine, metabolic, hematological disorders, gastrointestinal disorders, or immunodeficiency • Significant bleeding risk or coagulation disorders, history of gastrointestinal bleeding, which was clinically relevant • History or current hepatic disease • Requirement for anti-platelet or anti-coagulant medication

CIndU, chronic inducible urticaria; CSU, chronic spontaneous urticaria; HSS7, weekly Hives Severity Score; ISS7, weekly Itch Severity Score; UAS7, weekly Urticaria Activity Score

Table 2. Study endpoints

The trials are designed to demonstrate better efficacy of remibrutinib versus placebo in CSU with respect to the below endpoints:
Primary efficacy endpoints
• Change from baseline in UAS7 at Week 12 • Co-primary endpoints: Change from baseline in ISS7 and HSS7 at Week 12
Main secondary endpoints
• Proportion of participants achieving disease control (UAS7≤6) at Week 12 • Proportion of participants achieving complete absence of hives and itch (UAS7=0) at Week 12 • Early onset of disease control, defined as achievement of UAS7≤6 at Week 2 • Occurrence of treatment-emergent AEs and SAEs during the study

AEs, adverse events; HSS7, weekly Hives Severity Score; ISS7, weekly Itch Severity Score; SAE, serious AEs; UAS7, weekly Urticaria Activity Score

RESULTS

- The enrolment of the REMIX trials was initiated in Q4 2021, and the studies are expected to be completed in 2024
- A primary analysis may be conducted when all participants have completed Week 24 or discontinued early and when a minimum of 150 participants across both Phase 3 pivotal trials have completed the treatment period

CONCLUSIONS

- Remibrutinib, a novel BTK inhibitor, shows potential as a preferred oral treatment option for patients with CSU, inadequately controlled by second-generation H_1-antihistamines[5]
- The results of the REMIX trials will provide further evidence on the efficacy and safety of remibrutinib in CSU, compared to placebo

References

1. Zuberbier T, et al. *Allergy*. 2022; 77:734–766
2. van den Elzen MT, et al. *Clin Transl Allergy*. 2017;7:4
3. Hata D, et al. *J Exp Med*. 1998; 187:1235–1247
4. Angst D, et al. *J Med Chem*. 2020; 63:5102−5118
5. Maurer M, et al. *European Academy of Dermatology and Venereology*. 2021 (Oral Presentation #D3T01.3)

Disclosures

Marcus Maurer is or recently was a speaker and/or advisor for and/or has received research funding from Amgen, Allakos, Aralez, AstraZeneca, Celldex, FAES, Genentech, GI Innovation, Kyowa Kirin, Leo Pharma, Menarini, Novartis, Moxie, MSD, Roche, Sanofi, Third Harmonic, UCB and Uriach. **Ana Giménez-Arnau** reports roles as a Medical Advisor for Uriach Pharma, Sanofi and Genentech, Novartis, FAES, GSK, AMGEN, Thermo Fisher and has research grants supported by Uriach Pharma, Novartis and Instituto Carlos III- FEDER; she also participates in educational activities for Uriach Pharma, Novartis, Genentech, Menarini, LEO- PHARMA, GSK, MSD, Almirall, AVENE and Sanofi. **Sarbjit Saini** has received grant/research/clinical trial support from the National Institutes of Health, Novartis, Sanofi, Amgen and Regeneron, and is a consultant/advisory board member for Allakos, Granular Therapeutics, Novartis, Aquestive, Regeneron, Escient, Innate, Celltrion, and Sanofi. **Mark Lebwohl** is an employee of Mount Sinai and receives research funds from AbbVie, Amgen, Arcutis, Avotres, Boehringer Ingelheim, Dermavant Sciences, Eli Lilly, Incyte, Janssen Research & Development, LLC, Ortho Dermatologics, Regeneron, and UCB, Inc., and is a consultant for Aditum Bio, Almirall, AltruBio Inc., AnaptysBio, Arcutis, Aristea Therapeutics, Arrive Technologies, Avotres Therapeutics, BiomX, Boehringer Ingelheim, Bristol-Myers Squibb, Cara Therapeutics, Castle Biosciences, Corrona, Dermavant Sciences, Dr. Reddy's Laboratories, Evelo Biosciences, Evommune, Inc., Facilitatation of International Dermatology Education, Forte Biosciences, Foundation for Research and Education in Dermatology, Helsinn Therapeutics, Hexima Ltd., LEO Pharma, Meiji Seika Pharma, Mindera, Pfizer, Seanergy, and Verrica. **Gordon Sussman** has received research support from Aimmune, Amgen, Astra Zeneca, DBV technologies, Genentech, Kedrion S.p.A, Leo Pharma Inc., Novartis, Nuvo Pharmaceuticals Inc., Sanofi, Stallergenes, Merck, Schering Plough, Regeneron and ALK; is a medical advisor and/or has received payment for lectures from Merck, Novartis, CSL Behring, Pfizer, Anaphylaxis Canada, the Allergy Asthma and Immunology Society of Ontario and the Canadian Hereditary Angioedema Network. **El-Djouher Martzloff, Karine Lheritier, Artem Zharkov** and **Sibylle Haemmerle** are employees of Novartis Pharma AG, Basel, Switzerland.

Acknowledgements

All authors participated in the development of the poster for presentation. The authors wish to thank all investigators and patients involved in the trial. The authors thank Kate Killick (Novartis Ireland Ltd., Dublin) and Suparna Mukherjee (Novartis Healthcare Pvt. Ltd., Hyderabad) for editorial and medical writing support, and Hareesh Cheela for designing support (all Novartis Healthcare Pvt. Ltd., Hyderabad), which was as sponsored by Novartis Pharma AG, Basel, Switzerland in accordance with the Good Publication Practice (GPP3) guidelines (http:// www.ismpp.org/gpp3).

This investigation was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at: 6th GA²LEN Global Urticaria Forum (GUF) Congress, 7–8 December 2022, Berlin, Germany.

To download a copy of this poster, visit the web at: https://bit.ly/UKCGUF22

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission of the authors

Scan to download a copy of this poster