Poster 223

Jonathan A. Bernstein | bernstja@ucmail.uc.edu

Frequency of angioedema in chronic spontaneous urticaria patients: Report from the Urticaria Voices study

Jonathan A Bernstein,[1,2] Tonya A Winders,[3] Maria-Magdalena Balp,[4] Pallavi Saraswat,[5] Jessica McCarthy,[6] Tara Raftery,[7] Merin Kuruvilla,[6] Karsten Weller[8]

[1]Division of Allergy and Immunology, University of Cincinnati, Cincinnati, Ohio, USA; [2]Bernstein Allergy Group and Clinical Research Center, Cincinnati, Ohio, USA; [3]Global Allergy and Airways Patient Platform, Vienna, Austria; [4]Novartis Pharma AG, Basel, Switzerland; [5]Novartis Healthcare Pvt. Ltd, Hyderabad, India; [6]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA; [7]Novartis Ireland Ltd., Dublin, Ireland; [8]Institute of Allergology, Charité – Universitätsmedizin Berlin, Corporate Member of Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany

Scan to obtain:
• Poster

To download a copy of this poster, visit the web at:
https://bit.ly/UKCAAAAI
Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission of the author.

CONCLUSIONS

- Patients with CSU can experience significant burden due to angioedema despite treatment
- Patients reported higher rates of ever experiencing angioedema compared to physicians; however, the number of episodes of angioedema per year was similar
- Both patients and physicians reported being free of angioedema as an important treatment goal
- New therapies that are consistent with patients' and physicians' treatment goals of urticaria and angioedema control are needed

Poster presented at: The American Academy of Allergy, Asthma & Immunology Annual Meeting; February 23-26, 2024; Washington, DC.

INTRODUCTION

- Chronic spontaneous urticaria (CSU) is characterized by recurrent occurrences of itchy wheals (hives) with or without angioedema for more than 6 weeks[1]
- Approximately 50% of patients with CSU experience angioedema, which has been associated with increased disease severity and duration[2]
- The Urticaria Voices study aimed to assess the perceptions of patients with CSU, and physicians treating CSU on various aspects of disease management

OBJECTIVE

- The objective of this analysis was to describe the frequency and burden of angioedema as reported by patients and treatment goals from patients' and physicians' perspectives

METHODS

Study design and patients

- The Urticaria Voices was designed as a global (US, Canada, UK, Germany, France, Italy, and Japan) cross-sectional online quantitative survey including patients with CSU and physicians who treat CSU (**Figure 1**)

Figure 1. Global representation of patients with CSU and physicians treating CSU in Urticaria Voices study

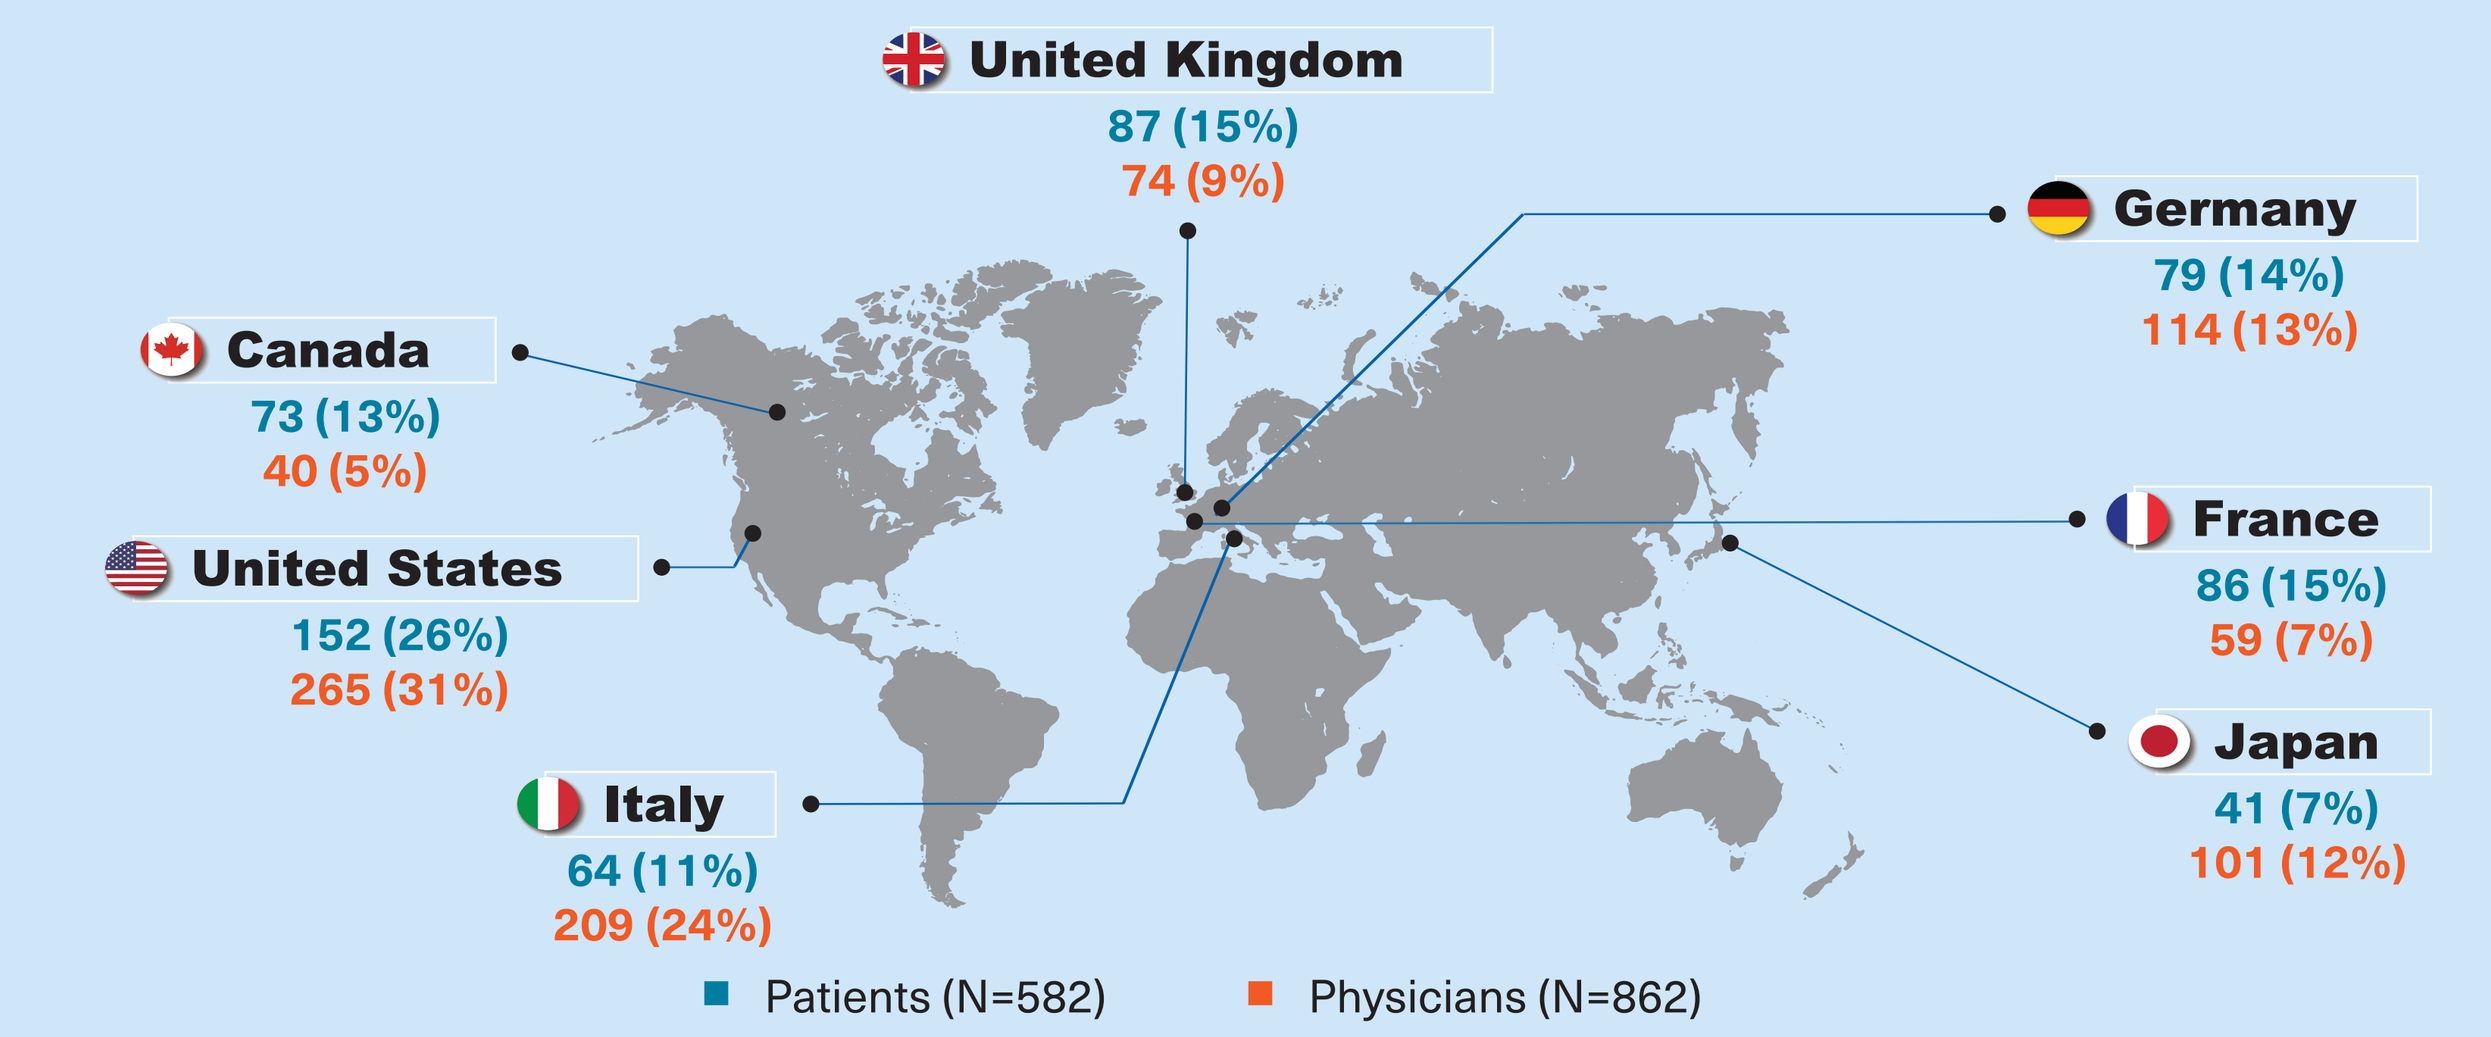
United Kingdom
87 (15%)
74 (9%)
Germany
79 (14%)
114 (13%)
Canada
73 (13%)
40 (5%)
France
86 (15%)
59 (7%)
United States
152 (26%)
265 (31%)
Japan
41 (7%)
101 (12%)
Italy
64 (11%)
209 (24%)
Patients (N=582)
Physicians (N=862)

Patient representation from each country is depicted by n (%).
n, number of patients from each country who participated in the study; N, total number of patients or physicians who participated in the study.

- Eligible adult patients (aged ≥18 years) had a self-reported clinician-provided diagnosis of CSU and were currently taking a prescribed treatment(s). Eligible physicians were medical specialists (dermatologists, allergists, or immunologists) who diagnose and treat patients with CSU
- Patients were recruited from online nationally representative panels and patient advocacy groups. Physicians were recruited from specialized online panels
- The 40-minute online patient survey and the 30-minute online physician survey were conducted between February 2022 and September 2022
- Patients survey assessed symptoms (itch, hives, angioedema), Urticaria Control Test (UCT), and treatments goals
- Physicians' survey assessed treatment, disease management, and goals
- Data by responder type were analyzed using descriptive statistics

RESULTS

- Overall, 582 patients with CSU (of which 36% had concomitant chronic inducible urticaria) and 862 physicians participated in the Urticaria Voices study
- The mean±SD age of CSU patients was 42.2±11.9 years. Most of the patients were female (62%). Patients reported a delay of approximately 2 years from the onset of first symptoms (mean±SD, 9.2±10.3 years) to receiving a diagnosis for their CSU (7.1±8.5 years) (**Figure 2**)

Figure 2. Patient demographics and disease characteristics

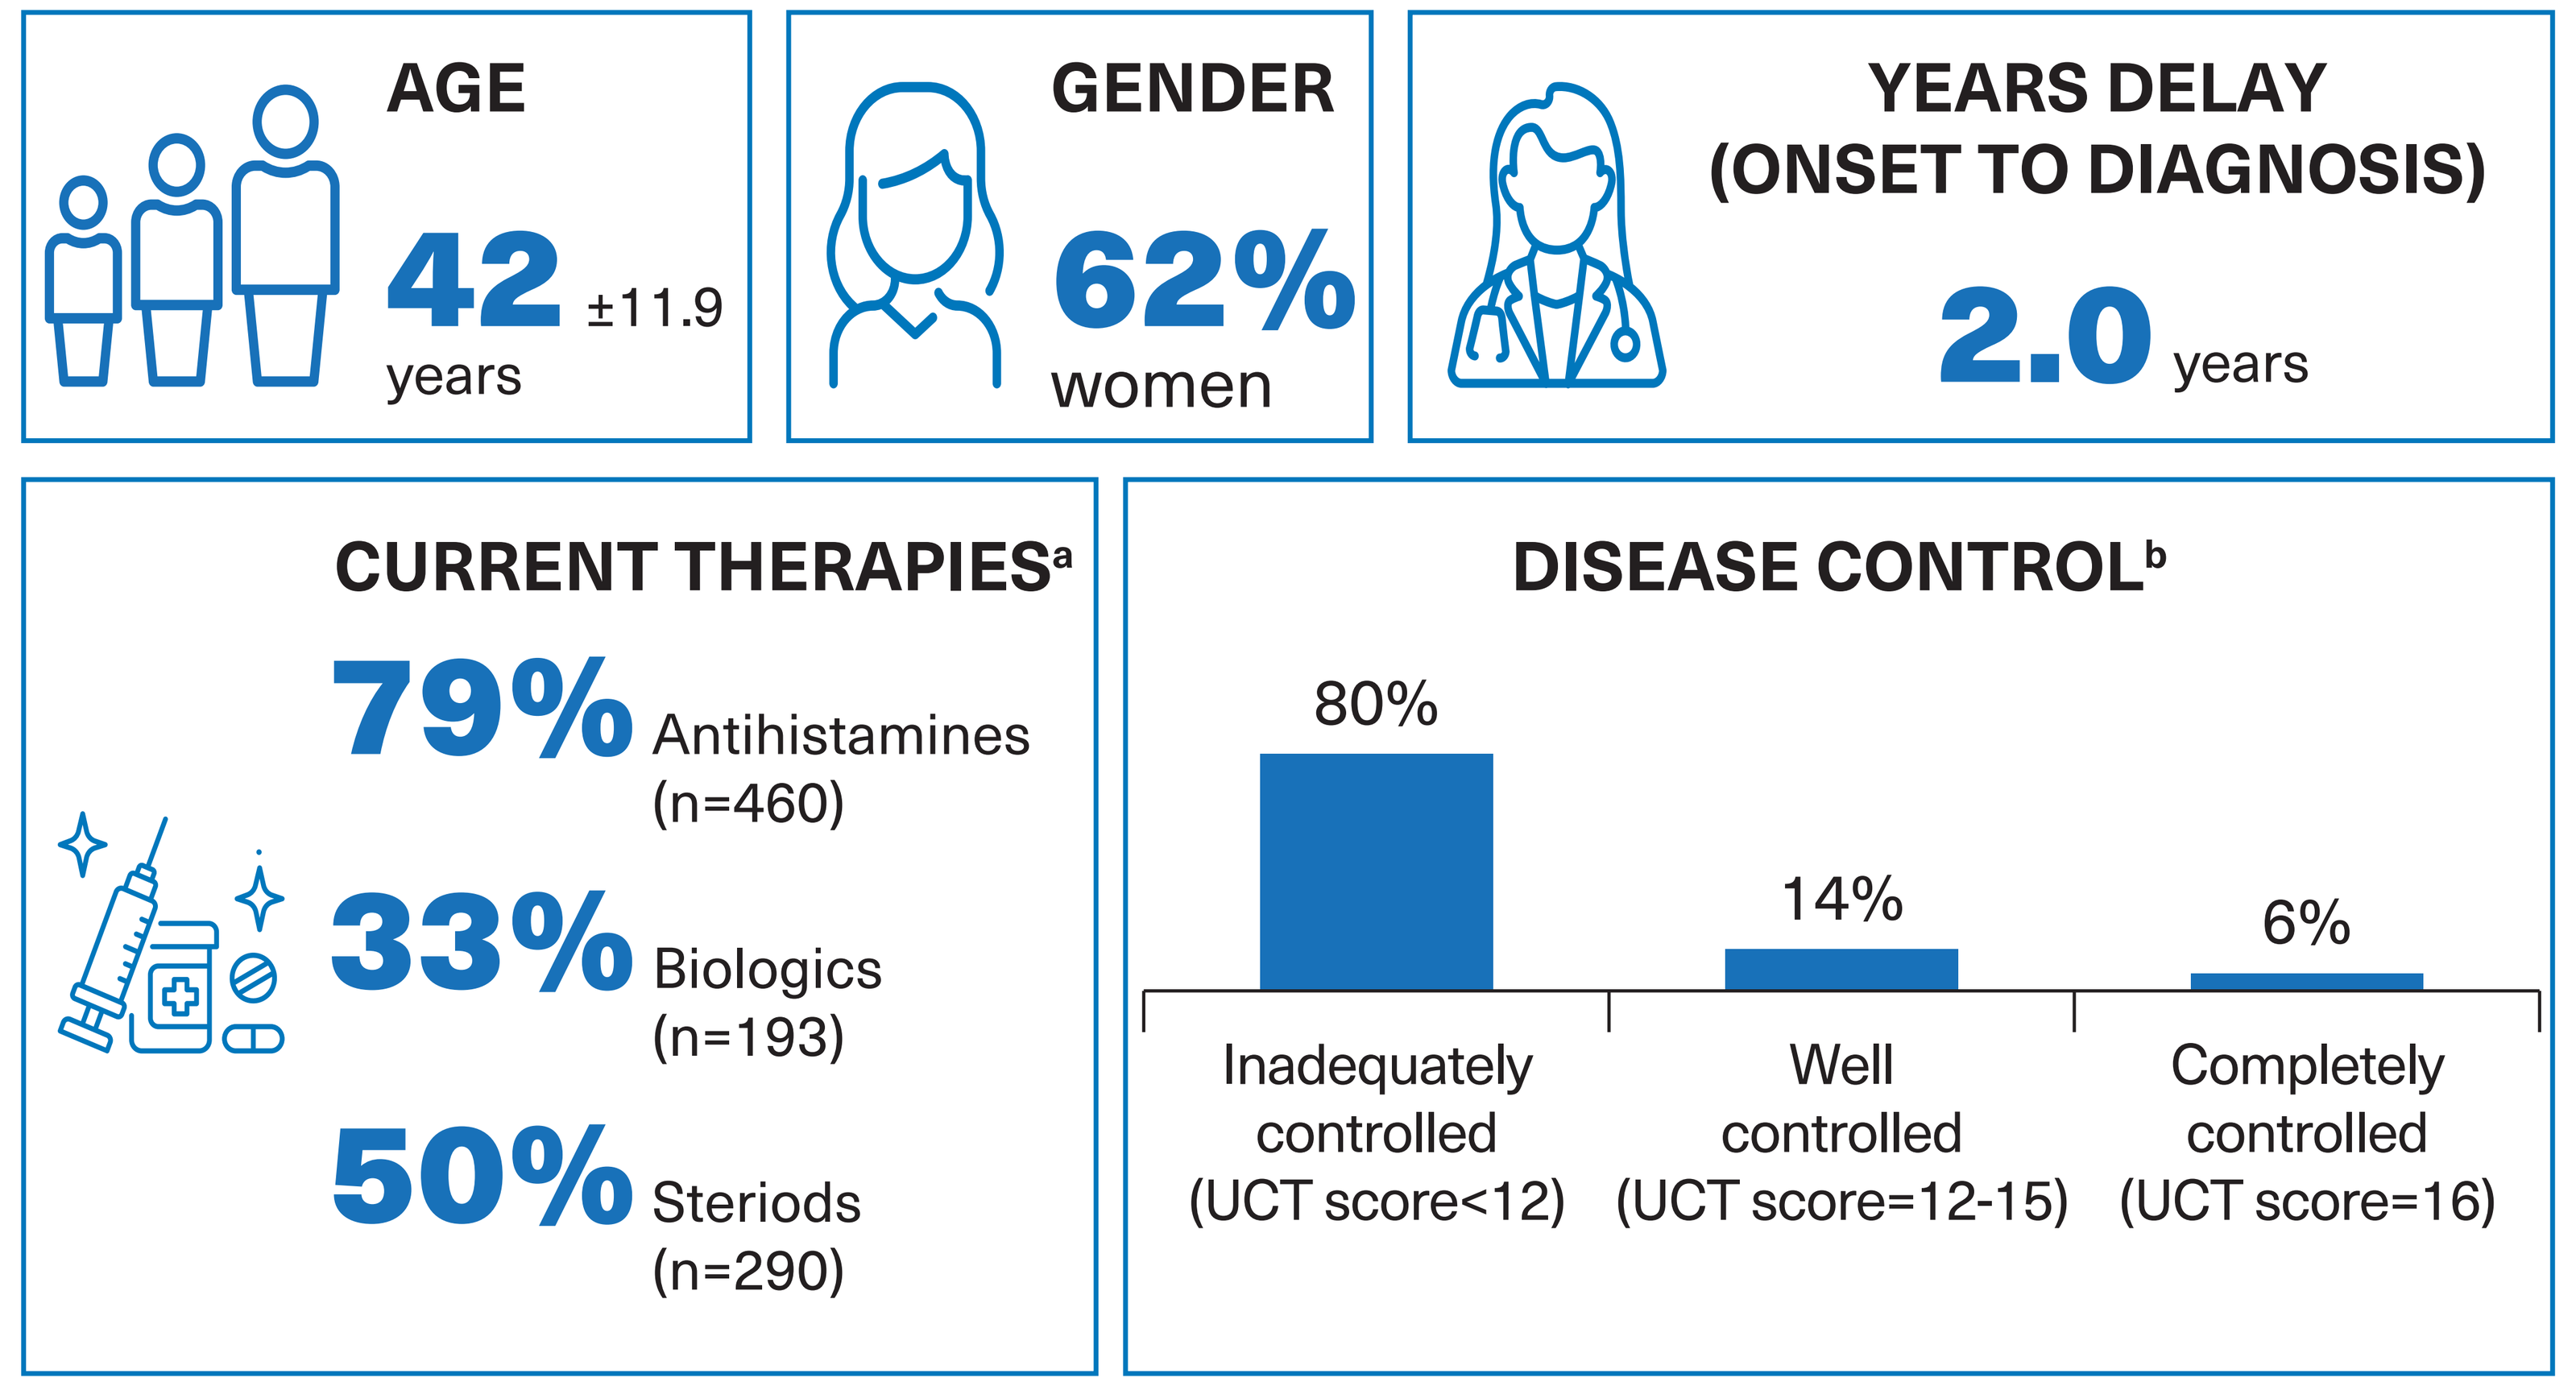
AGE
42 ±11.9
years
GENDER
62%
women
YEARS DELAY
(ONSET TO DIAGNOSIS)
2.0 years
CURRENT THERAPIES[a]
79% Antihistamines
(n=460)
33% Biologics
(n=193)
50% Steriods
(n=290)
DISEASE CONTROL[b]
80%
14%
6%
Inadequately controlled (UCT score<12)
Well controlled (UCT score=12-15)
Completely controlled (UCT score=16)

Age is represented as mean±SD; [a]Patients may have been on combination therapies at the time of survey; [b]Assessed over the previous 4 weeks.
CSU, chronic spontaneous urticaria; n, number of patients; UCT, Urticaria Control Test.

- Globally, patients reported higher rates of ever experiencing angioedema and number of angioedema episodes per year compared to physicians (**Figure 3**)
- Both physicians (69%) and patients (50%) reported being "free of angioedema" as an important treatment goal (**Figure 3**)

Figure 3. Frequency of angioedema episodes in patients with CSU, and CSU treatment goals – Patients' and physicians' perception

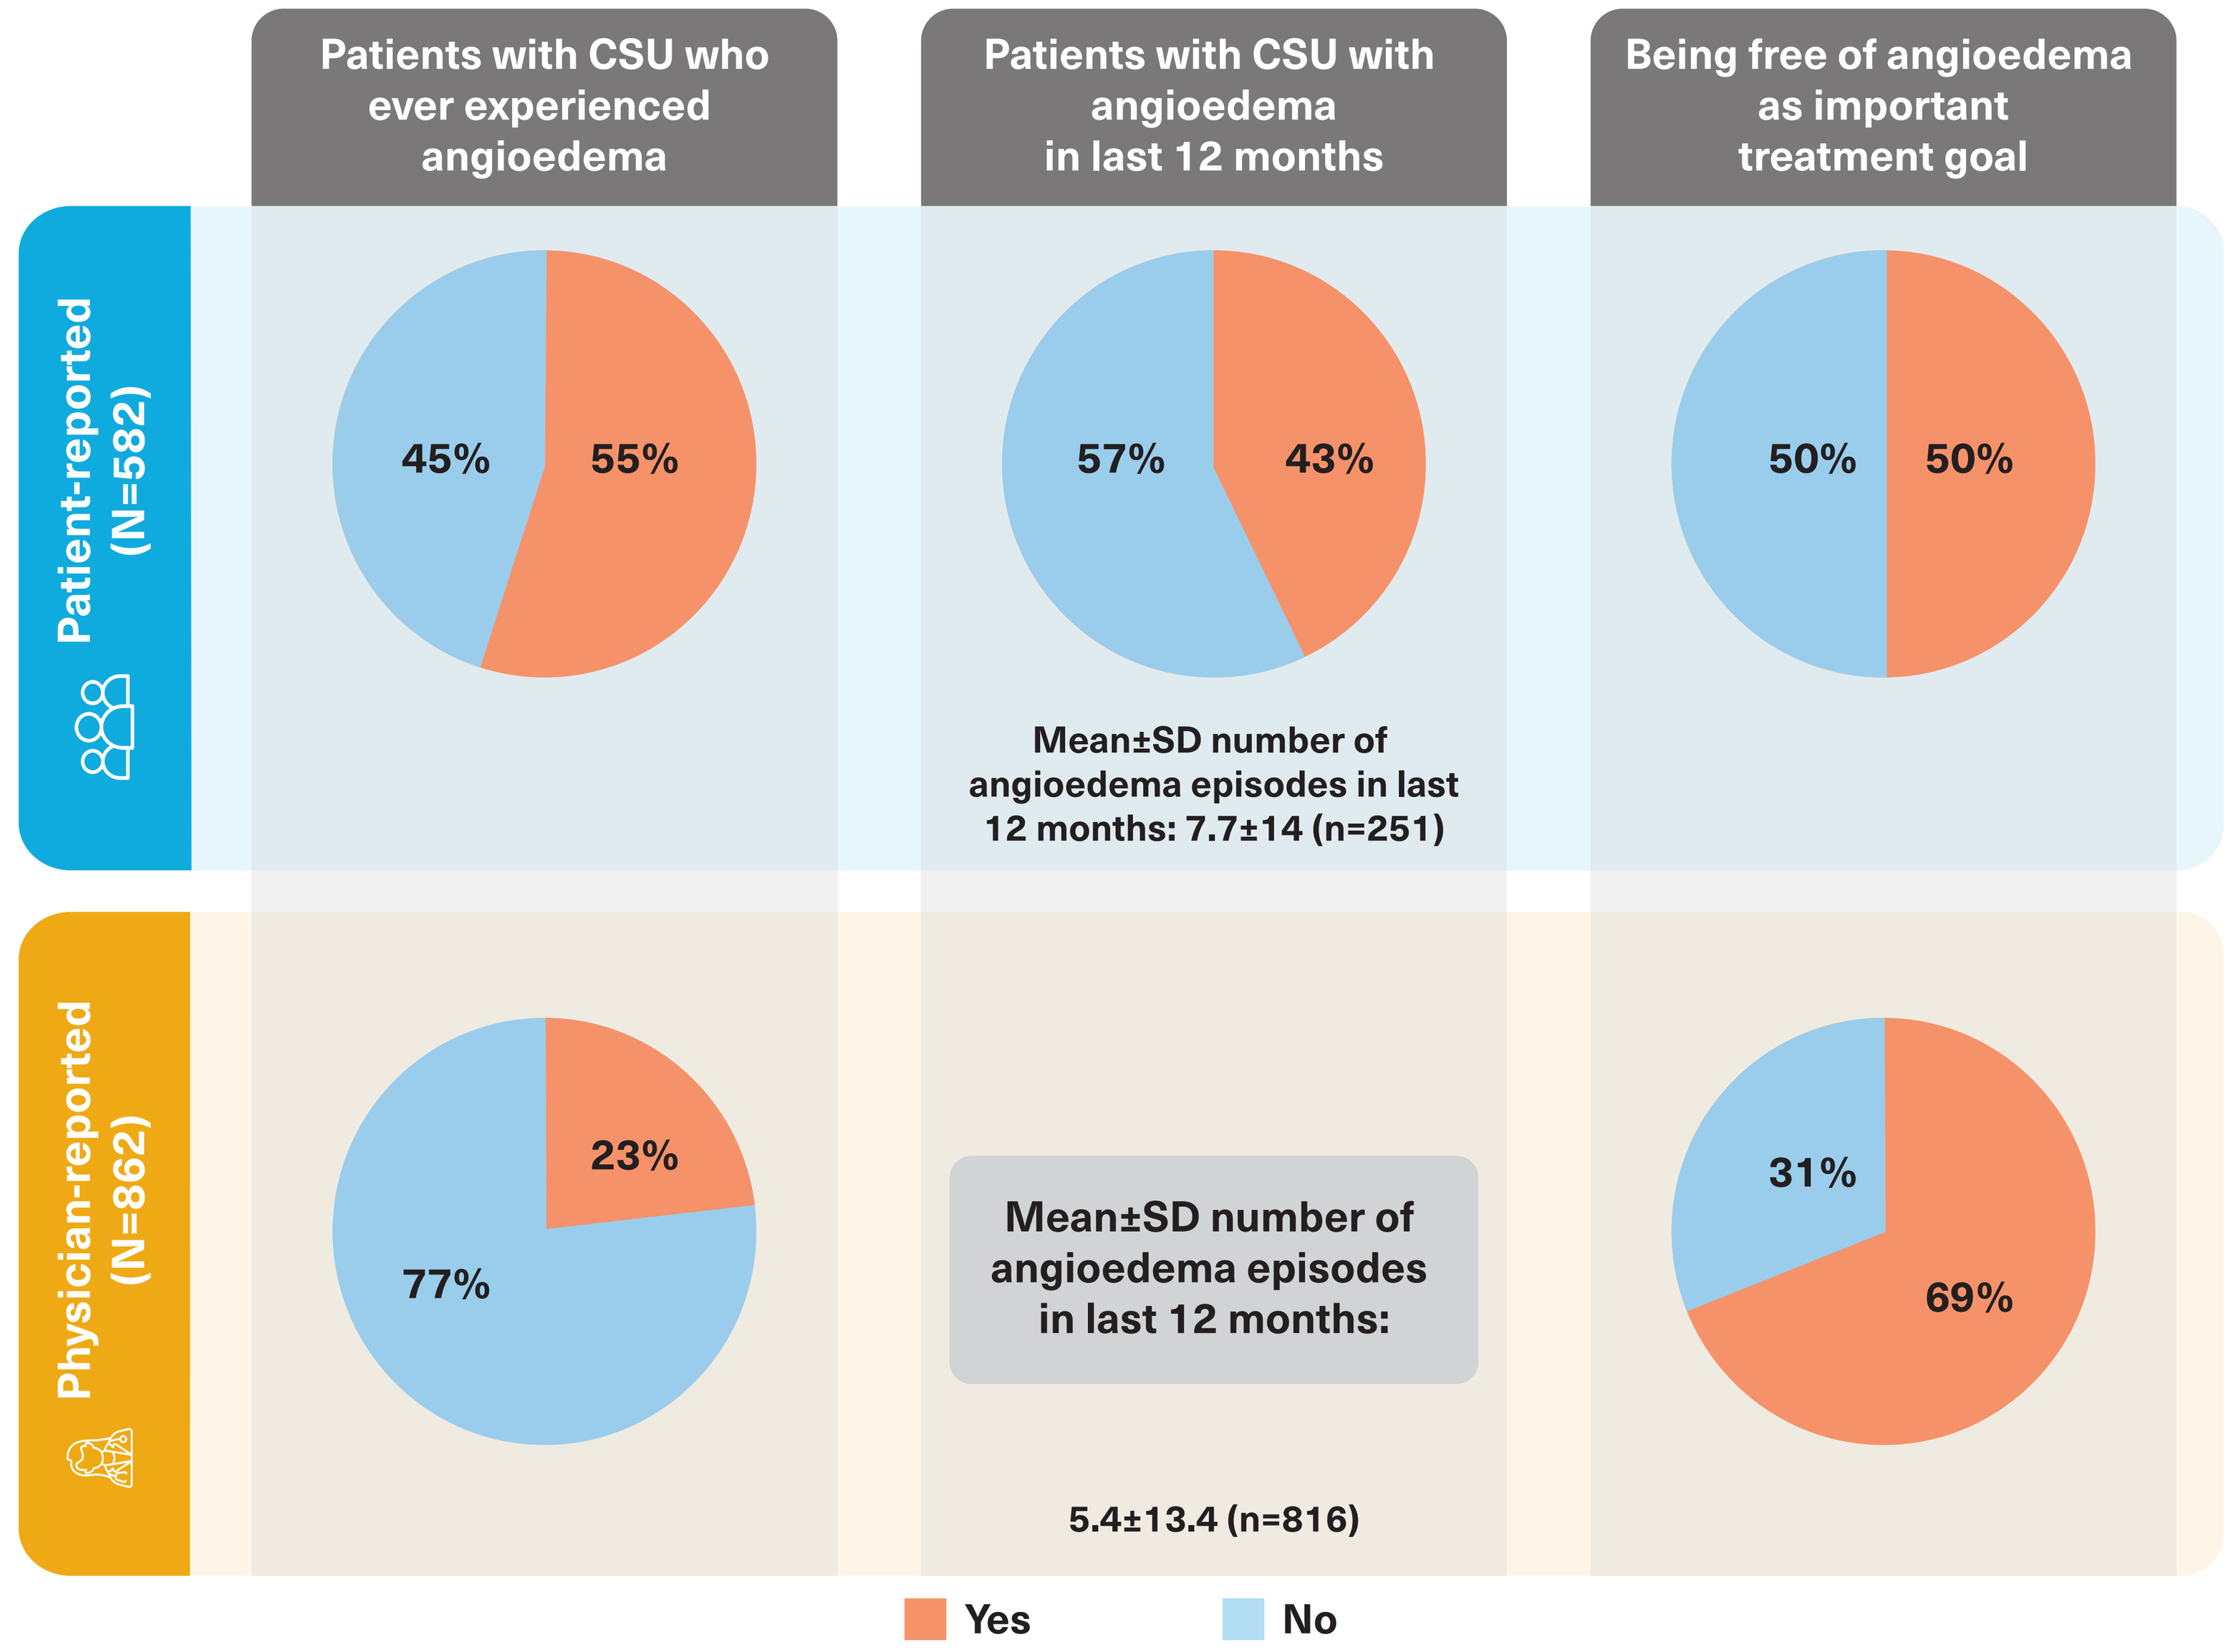
Patients with CSU who ever experienced angioedema
Patients with CSU with angioedema in last 12 months
Being free of angioedema as important treatment goal
Patient-reported (N=582)
45%
55%
57%
43%
Mean±SD number of angioedema episodes in last 12 months: 7.7±14 (n=251)
50%
50%
Physician-reported (N=862)
23%
77%
Mean±SD number of angioedema episodes in last 12 months:
5.4±13.4 (n=816)
31%
69%
Yes
No

CSU, chronic spontaneous urticaria; N, total number of physicians or patients with CSU; n, number of patients with angioedema episodes in last 12 months; SD, standard deviation.

- Generally, a higher proportion of patients reported angioedema ever and the mean number of angioedema episodes per year was higher than physician reported data, regardless of the country (**Table 1**)
- Number of episodes of angioedema reported in the last 12 months were broadly aligned between patients and physicians (**Table 1**)

Table 1. Country-wide data on frequency and number of angioedema episodes in patients with CSU, and CSU treatment goal as reported by patients and physicians

	Ever experienced angioedema (%)		Angioedema episodes in last 12 months[a] (mean±SD)		Treatment goal: Be free of angioedema (%)	
	Patients (N=582)	Physicians (N=862)	Patients (N=251)	Physicians (N=862)	Patients (N=582)	Physicians (N=862)
Italy	22	23	2.38±1.69	4.85±7.06	48	73
Canada	49	17.9	18.41±30.49	3.67±4.99	62	78
France	58	21.27	7.62±11.33	4.04±3.91	51	66
Germany	59	24.68	5.81±15.95	7.19±17.62	39	71
Japan	37	17.31	5.64±6.30	3.66±5.87	46	55
UK	62	28.92	4.74±3.47	7.14±14.04	53	65
USA	67	22.58	7.19±6.96	5.7±18.05	49	72

[a]Among patients who reported angioedema in the last 12 months.
CSU, chronic spontaneous urticaria; N, total number of patients or physicians.

- Overall, 44% of patients with currently adequately controlled CSU (UCT≥12) and 57% with inadequately controlled CSU (UCT<12) reported that they had experienced angioedema ever in their lifetime
- Of patients currently on H1-antihistamines (79%; 460/582), 47% (215/460) reported angioedema in the last 12-months (mean±SD, 7.7±15 episodes)
- A proportion of 34% of patients reported pain at the site of angioedema as a symptom of CSU

Strengths and limitations of the Urticaria Voices study

- This was a multinational real-world evidence study on the perceived burden of CSU, captured from the perspective of patients with CSU and treating physicians with a large sample size supporting generalizability of the results
- Online surveys were limited to patients who had access to internet and as with all surveys were subjected to recollection bias
- The patient and physician populations were not matched (physicians surveyed were not treating the patients surveyed)

Disclosures

Jonathan A. Bernstein reports grants from Novartis, Astra Zeneca, Sanofi-Regeneron, Amgen, Roche, Allakos, Celldex, CSL Behring, Takeda/Shire, Biocryst, Pharming, Ionis, Biomarin, and Genentech outside the submitted work; personal fees from Novartis, Astra Zeneca, Sanofi-Regeneron, Amgen, Roche, Allakos, Celldex, CSL Behring, Takeda/Shire, Biocryst, Pharming, Ionis, Biomarin, and Genentech outside of the submitted work. **Tonya Winders** receives funds from unbranded disease awareness & education from Novartis, AstraZeneca, Sanofi-Regeneron, Amgen, Roche & Genentech outside of the submitted work and was an employee of the Allergy and Asthma Network at the time of study conduct. **Maria-Magdalena Balp** is an employee of Novartis Pharma AG, Basel, Switzerland. **Pallavi Saraswat** is an employee of Novartis Healthcare Pvt. Ltd, Hyderabad, India. **Jessica McCarthy** and **Merin Kuruvilla** are employees of Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA. **Tara Raftery** is an employee of Novartis Ireland Ltd, Dublin, Ireland. **Karsten Weller** reports grants from Novartis and Takeda outside of the submitted work and personal fees from Biocryst, Biomarin, CSL Behring, Novartis, Moxie, Takeda outside of the submitted work.

Acknowledgments

All authors participated in the development of the poster for presentation. The authors wish to thank all investigators and patients involved in the trial. The authors thank **Pedro A Laires** (former employee of Novartis Pharma AG, Basel, Switzerland) for his contribution to the study and **Mihai Dricu** (Ipsos SA, Basel, Switzerland) for statistical analysis. The authors also thank **Sahaja Banda** and **Suparna Mukherjee** for editorial and medical writing support and **Srinivas Kanchipati** for the design support (all Novartis Healthcare Pvt. Ltd., Hyderabad, India), which was funded by Novartis Pharma AG, Basel, Switzerland, in accordance with the Good Publication Practice (GPP3) guidelines (http://www.ismpp.org/gpp3).

Funding

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

References

1. Zuberbier T, et al. *Allergy*. 2022;77(3):734–766.
2. Sussman G, et al. *Allergy*. 2018;73(8):1724–1734.